The Effectiveness of Darbepoetin alfa Administered Every 3 Weeks on Clinical Outcomes in Elderly Patients With Chemotherapy-Induced Anemia

Ralph Boccia,[1] Peter Silberstein,[2] Simon Tchekmedyian,[3] Dianne Tomita,[4] Greg Rossi,[4] and Greg Otterson[5]

[1]Georgetown University Hospital, Washington, DC; [2]Creighton University, Omaha, NE; [3]Pacific Shores Medical Group, Huntington Beach, CA; [4]Amgen Inc., Thousand Oaks, CA; [5]Ohio State University, Columbus, OH

INTRODUCTION

- Chemotherapy-induced anemia (CIA) and associated fatigue are common in patients with cancer[1] and can be effectively alleviated by increasing hemoglobin (Hb) levels with erythropoietic agents such as darbepoetin alfa (Aranesp®) and epoetin alfa (Procrit®).[2,3]
- Evidence-based guidelines recommend that Hb levels are maintained between 11 and 13 g/dL during erythropoietic therapy.[4-6]
- Darbepoetin alfa therapy is effective at alleviating the symptoms associated with CIA when administered once every week,[7] every 2 weeks,[8,9] or every 3 weeks (Q3W).[10,11]
- Chemotherapy is usually administered Q3W. Administration of darbepoetin alfa Q3W may enable the synchronization of anemia therapy with chemotherapy, potentially offering added convenience to patients and their caregivers.
- We analyzed data from patients ≥ 65 years old and < 65 years old who were enrolled in a multicenter, open-label, 16-week study of patients with CIA. We evaluated the effectiveness of darbepoetin alfa 300 mcg Q3W in achieving and maintaining Hb levels within the range recommended by evidence-based guidelines and assessed patient-reported symptoms of fatigue using the Functional Assessment of Cancer Therapy-Fatigue (FACT-F) scale.

STUDY OBJECTIVE

- The primary objective of this interim analysis was to assess the effectiveness of darbepoetin alfa administered at 300 mcg Q3W in achieving and maintaining Hb levels between 11.0 and 13.0 g/dL, consistent with the therapeutic objectives of anemia treatment defined by current evidence-based guidelines.

PATIENTS AND METHODS

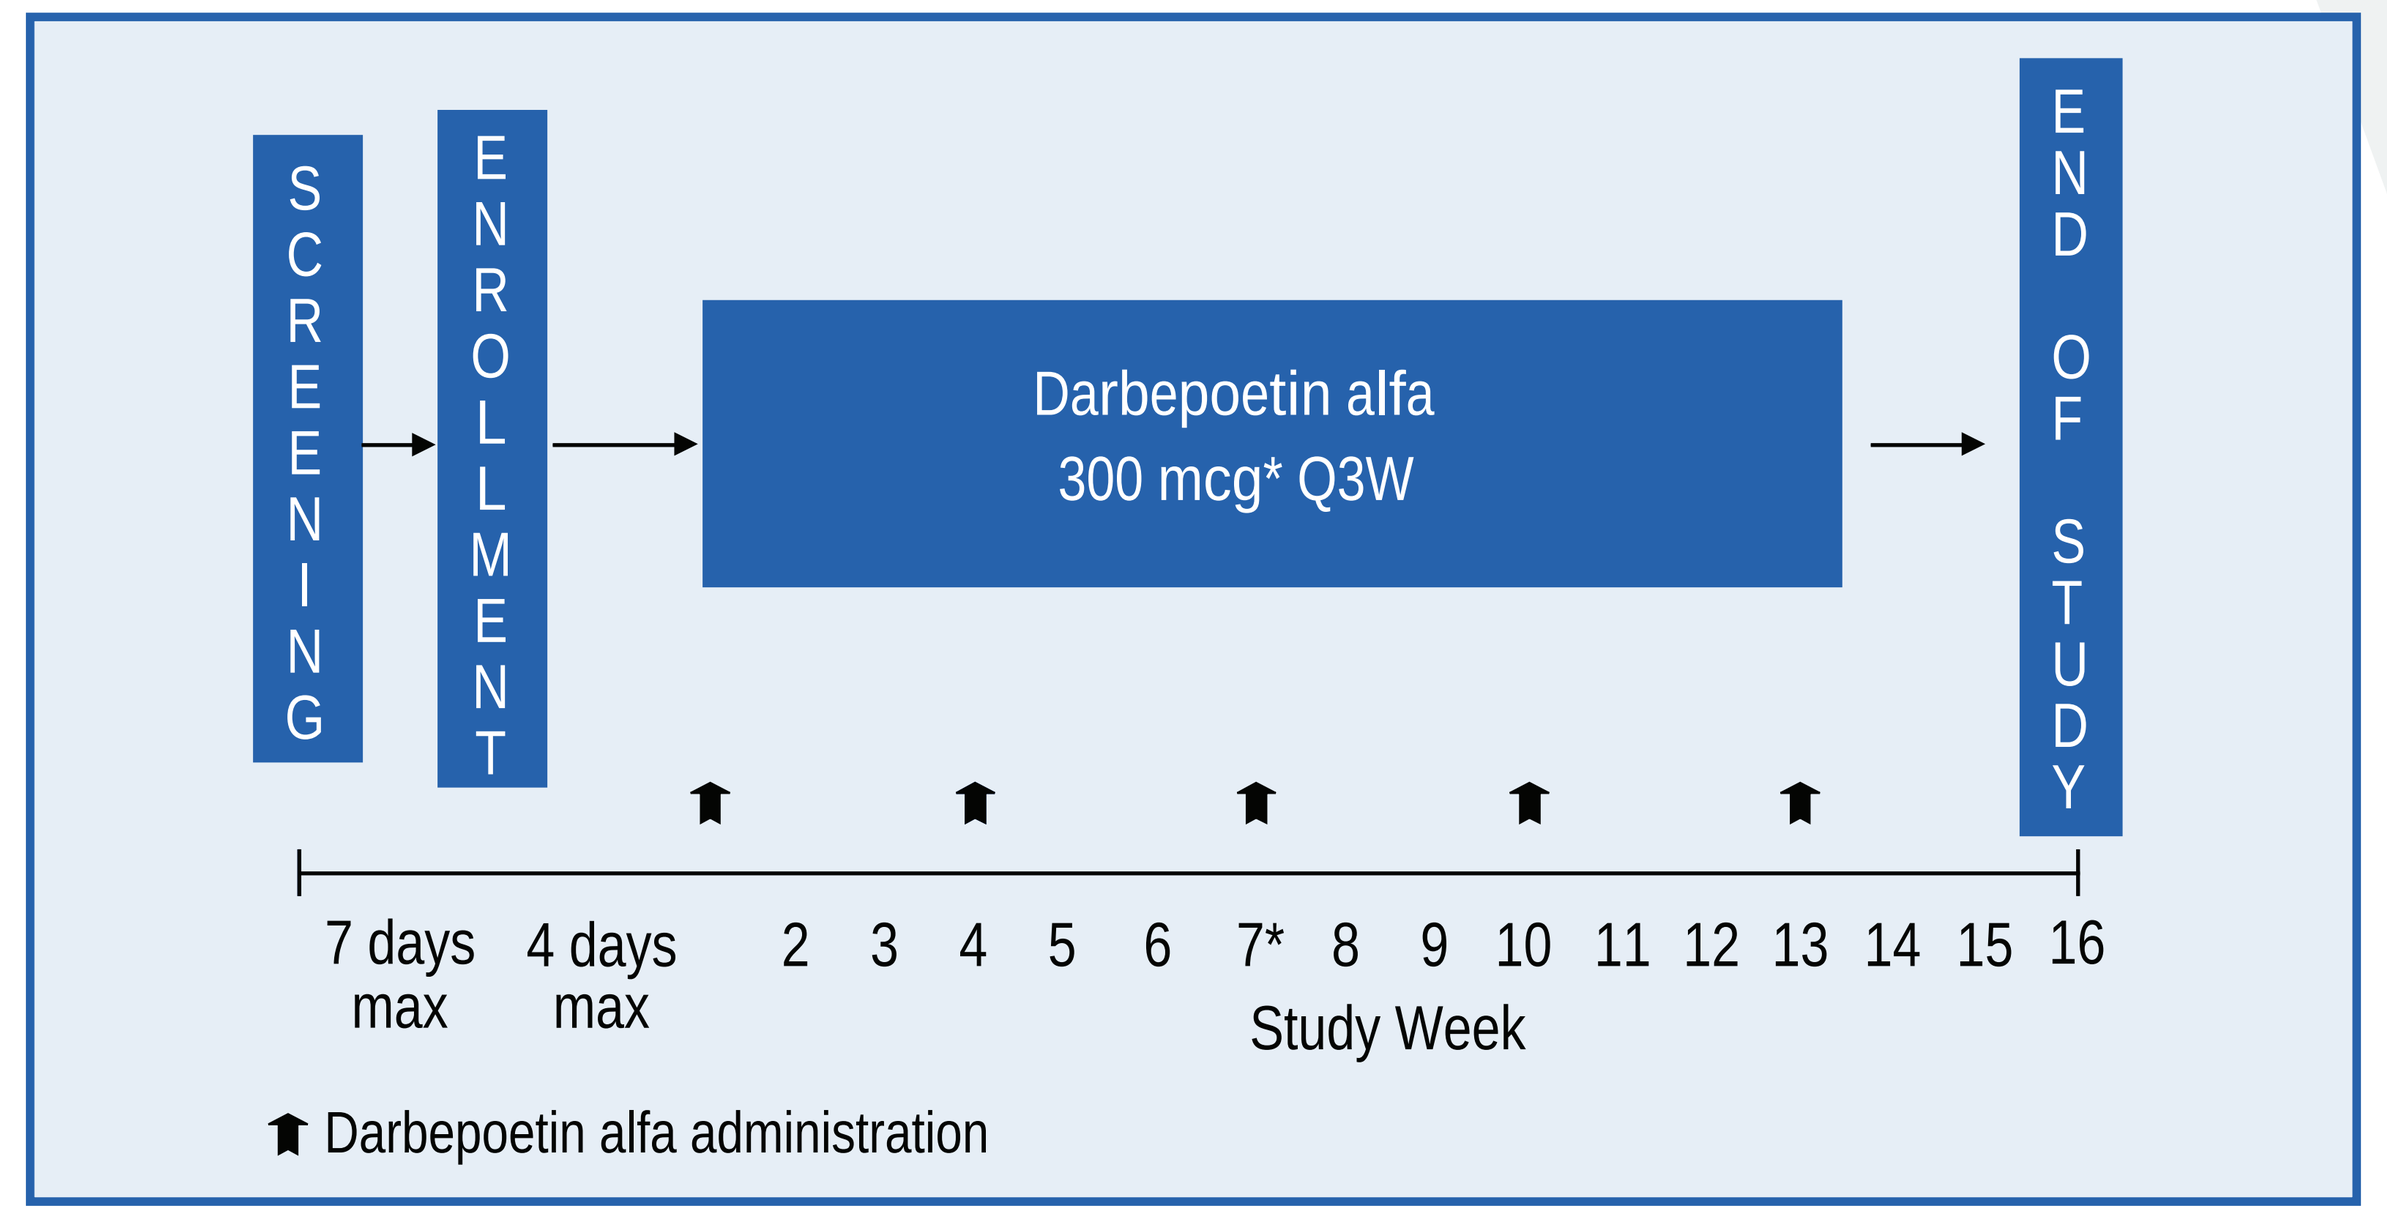
SCREENING
ENROLLMENT
Darbepoetin alfa
300 mcg* Q3W
END OF STUDY
7 days max
4 days max
2 3 4 5 6 7* 8 9 10 11 12 13 14 15 16
Study Week
Darbepoetin alfa administration

*Dose escalation was permitted after 6 weeks (2 doses). The dose was escalated to 500 mcg Q3W if the Hb remained below 10.0 g/dL and the increase in Hb from baseline was less than 1.0 g/dL. The dose was not escalated if the Hb was within the Hb target range. Darbepoetin alfa was withheld when Hb levels reached or exceeded 13.0 g/dL.

PATIENTS AND METHODS *(continued)*

Key Eligibility Criteria

- Anemic (Hb < 11 g/dL) at screening
- ≥ 18 years old
- Non-myeloid malignancy
- At least 8 additional weeks of chemotherapy
- Adequate liver and renal function
- No red blood cell (RBC) transfusions in the 2 weeks before screening
- No erythropoietic therapy in the 4 weeks before screening

Study Endpoints

- The primary endpoint was the percentage of patients who achieved and maintained Hb levels within the range of 11 to 13 g/dL as recommended by the current evidence-based guidelines
- Secondary endpoints included the percentage of patients who had a hematopoietic response, the percentage of patients that required RBC transfusions, and the change in FACT-F scores from baseline to the end of study

Statistical Analysis

- We analyzed data from 1225 patients who could have completed 16 weeks of treatment and received at least one dose of darbepoetin alfa
- Patients were stratified by age (≥ 65 years old and < 65 years old) and baseline Hb (< 10 g/dL and ≥ 10 g/dL) at screening
- Two approaches were used to analyze Hb-based endpoints:
 - Intent-to-treat (ITT) analysis: missing Hb values or values within 28 days of an RBC transfusion were excluded and imputed using the last value carried forward (LVCF) approach
 - Available data approach: missing Hb values were not imputed, and Hb values within 28 days of an RBC transfusion were excluded and not imputed
- Patient-reported outcomes were assessed using the FACT-Fatigue scale using available data at week 7 and at the end of the study

REFERENCES

1. Groopman JE, Itri LM. *J Natl Cancer Inst.* 1999;91(19):1616-1634.
2. Littlewood TJ, Bajetta E, Nortier JW, Vercammen E, Rapoport B. *J Clin Oncol.* 2001;19(11):2865-2874.
3. Vansteenkiste J, Pirker R, Massuti B, Barata F, Font A, Fiegl M, et al. *J Natl Cancer Inst.* 2002;94(16):1211-1220.
4. Rizzo JD, Lichtin AE, Woolf SH, Seidenfeld J, Bennett CL, Cella D, et al. *J Clin Oncol.* 2002;20(19):4083-4107.
5. Rodgers GM. In: http://www.nccn.org/professionals/physician_gls/f_guidelines.asp. Accessed April 18, 2005.
6. Bokemeyer C, Aapro MS, Courdi A, Foubert J, Link H, Osterborg A, et al. *Eur J Cancer.* 2004;40(15):2201-2216.
7. Glaspy JA, Jadeja JS, Justice G, Kessler J, Richards D, Schwartzberg L, et al. *Br J Cancer* 2002;87:268-276.
8. Patton J, Reeves T, Wallace J. *Oncologist.* 2004;9(4):451-458.
9. Schwartzberg L, Shiffman R, Tomita D, Stolshek B, Rossi G, Adamson R. *Clin Ther.* 2003;25(11):2781-2796.
10. Kotasek D, Steger G, Faught W, Underhill C, Poulsen E, Colowick AB, et al. *Eur J Cancer.* 2003;39(14):2026-2034.
11. Glaspy J. *Best Pract Res Clin Haematol.* 2005;18(3):407-416.
12. Cella D, Eton DT, Lai JS, Peterman AH, Merkel DE. *J Pain Symptom Manage.* 2002;24(6):547-561.

RESULTS

Baseline Demographics and Disease Characteristics

Characteristic	Elderly Patients (≥ 65 years old) (n = 584)	Younger Patients (< 65 years old) (n = 641)
Sex - n (%)		
Women	300 (51)	449 (70)
Race - n (%)		
White	497 (85)	472 (74)
Black	57 (10)	102 (16)
Other	30 (5)	67 (10)
Age (years)		
Mean (SD)	73.7 (5.6)	52.0 (9.1)
Primary tumor type - n (%)		
Gastrointestinal	141 (24)	137 (21)
Hematologic[a]	107 (18)	62 (10)
Breast	94 (16)	260 (41)
Lung	92 (16)	77 (12)
Genitourinary	80 (14)	25 (4)
Gynecologic	32 (5)	36 (6)
Other[b]	38 (7)	44 (7)
Mean (SD) baseline Hb (g/dL)	10.2 (0.7)	10.1 (0.8)
Mean (SD) baseline FACT-F	28.0 (12.5)	27.1 (12.1)

[a]May include acute or chronic lymphocytic leukemia, non-Hodgkin's lymphoma, Hodgkin's disease, multiple myeloma or other lymphoma

Achievement and Maintenance of Hemoglobin Levels

	Elderly Patients (≥ 65 years old) (n = 584)	Younger Patients (< 65 years old) (n = 641)
Achievement of target Hb (≥ 11 g/dL)		
K-M percentage (95% CL) of patients	92 (85, 99)	86 (83, 89)
Time to target Hb, K-M median, weeks (95% CL) [n]	4.0 (4.0, 5.0) [583]	4.0 (4.0, 5.0) [640]
Mean (95% CL) Hb after achieving target (g/dL) [n]	11.5 (11.5, 11.6) [460]	11.6 (11.5, 11.6) [509]
Patients maintaining Hb concentration after achieving target[a] - n (%)		
< 11.0 g/dL	112 (24)	125 (25)
11.0 to 13.0 g/dL	330 (72)	369 (72)
> 13.0 g/dL	18 (4)	15 (3)

[a]Based on number of patients who achieved the target Hb
K-M = Kaplan-Meier

Hemoglobin Endpoints

	Elderly Patients (≥ 65 years old) (n = 584)	Younger Patients (< 65 years old) (n = 641)
Change in Hb from baseline to week 16		
Mean (95% CL) (LVCF approach) [n]	1.1 (0.9, 1.2) [551]	1.1 (1.0, 1.3) [590]
Mean (95% CL) (available data approach) [n]	1.5 (1.3, 1.6) [364]	1.5 (1.3, 1.6) [404]
Patients with a hematopoietic response[a]		
Crude percent (95% CL)	61 (57, 65)	65 (61, 69)

[a] ≥ 2.0 g/dL rise in hemoglobin from baseline or Hb ≥ 12.0 g/dL in the absence of a transfusion

Time to Target Hemoglobin

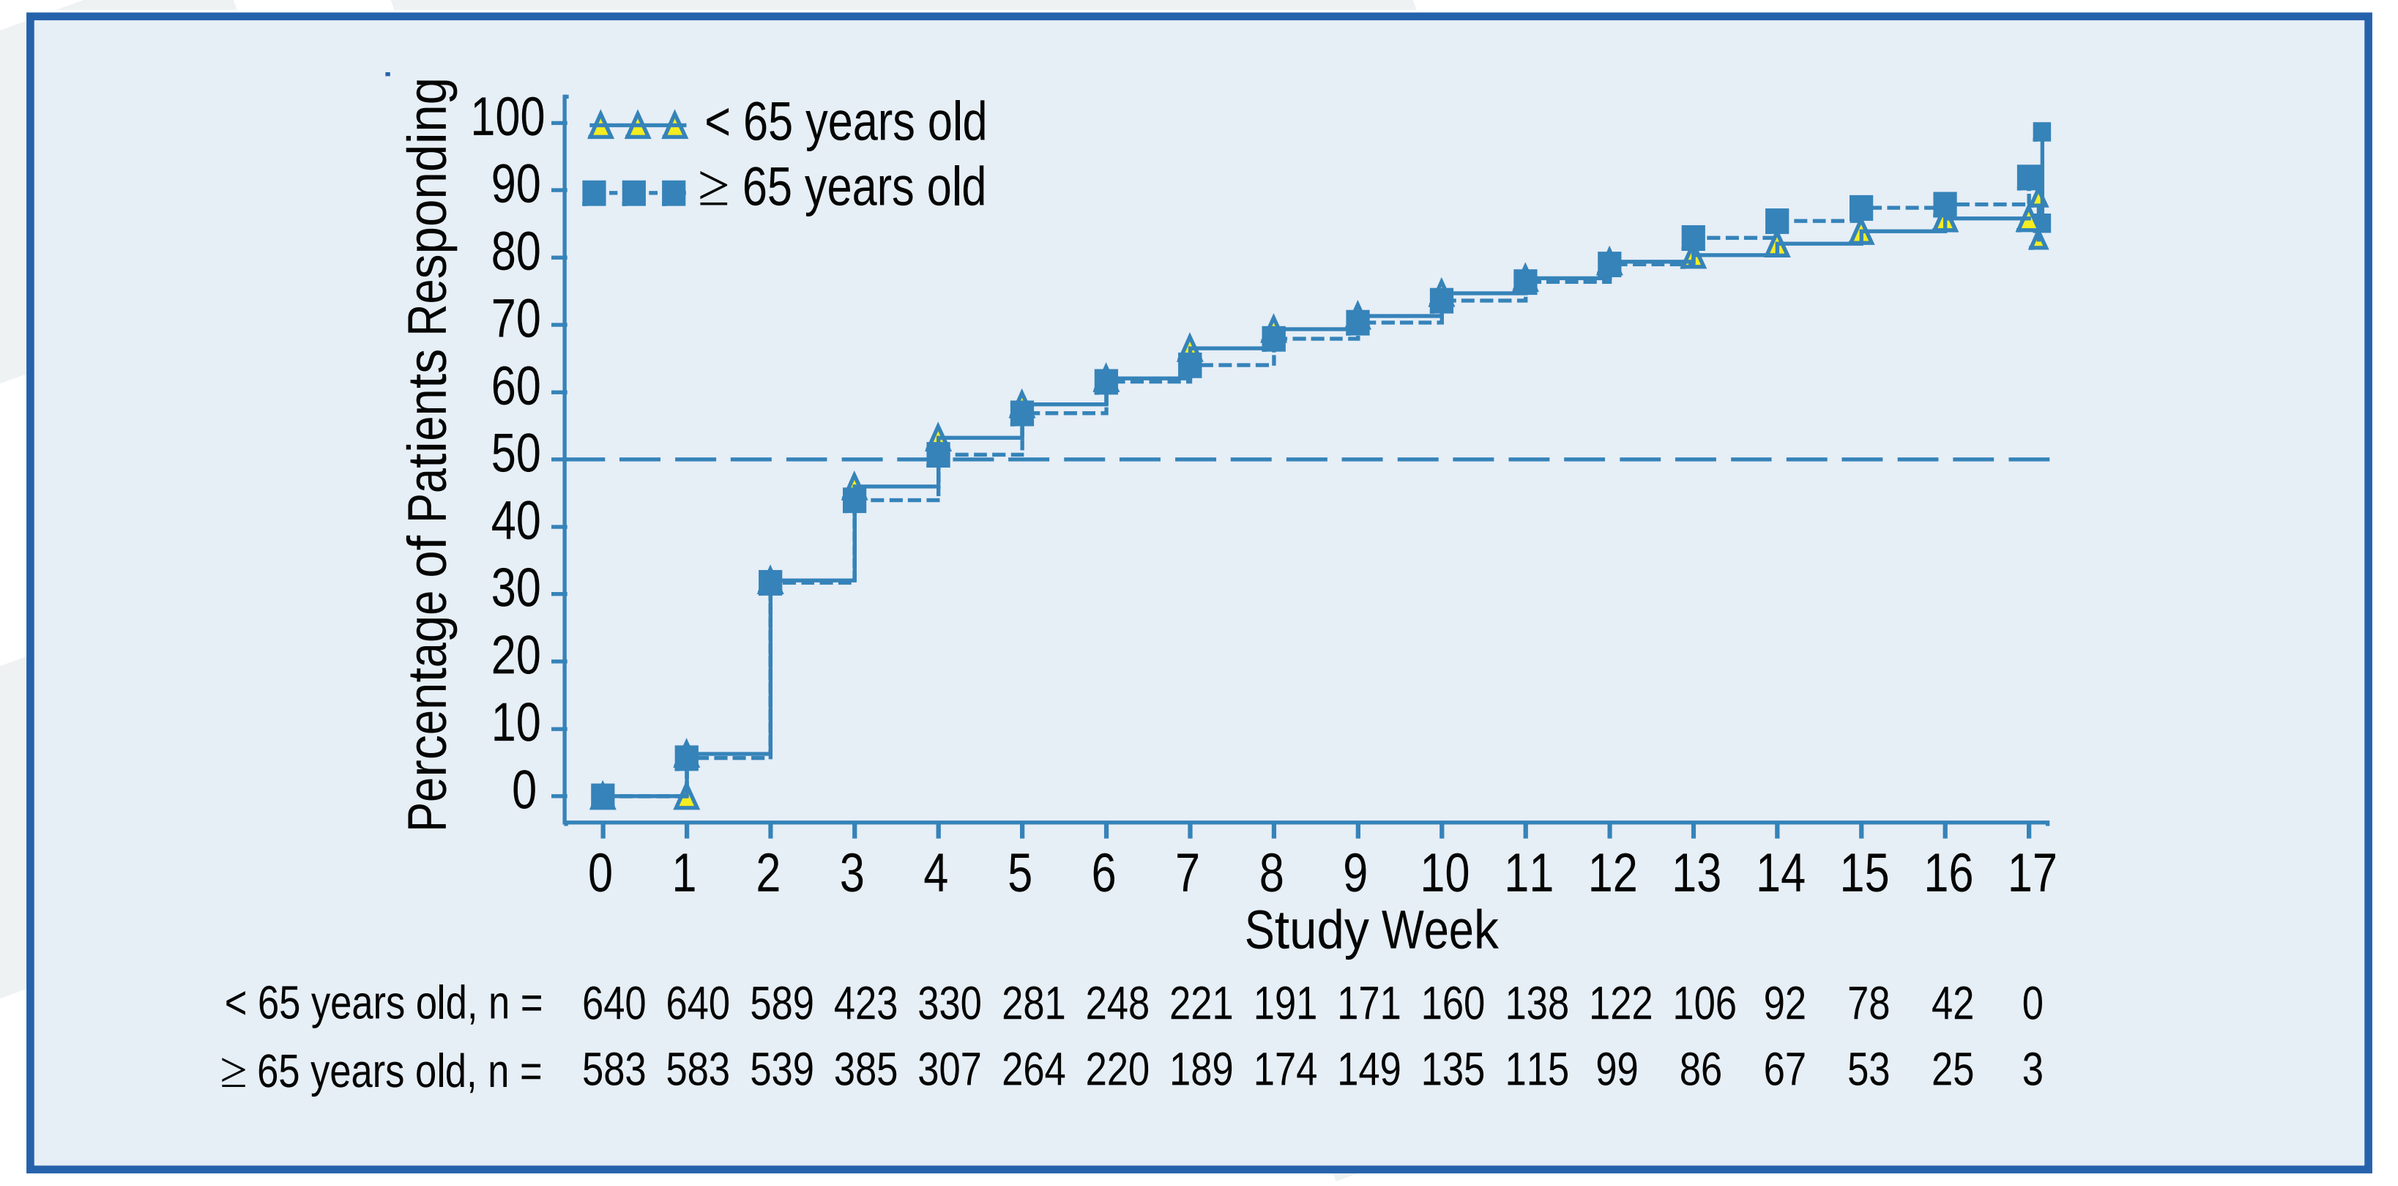
Percentage of Patients Responding
100
90
80
70
60
50
40
30
20
10
0
< 65 years old
≥ 65 years old
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17
Study Week
< 65 years old, n = 640 640 589 423 330 281 248 221 191 171 160 138 122 106 92 78 42 0
≥ 65 years old, n = 583 583 539 385 307 264 220 189 174 149 135 115 99 86 67 53 25 3

Percentage of Patients who Required Transfusions

	Elderly Patients (≥ 65 years old) (n = 584)	Younger Patients (< 65 years old) (n = 641)
Patients with RBC transfusions from week 1 to end of study (n)	582	641
K-M percent (95% CL)[a]	25 (21, 29)	26 (22, 29)
Crude percent (95% CL)	23 (20, 27)	24 (21, 28)
Patients with RBC transfusions from week 5 to end of study (n)	554	612
K-M percent (95% CL)[a]	19 (16, 22)	21 (17, 24)
Crude percent (95% CL)	17 (14, 20)	20 (16, 23)

[a]Determined from the Kaplan-Meier estimate

Darbepoetin alfa Q3W Reduced the Percentage of Patients That Required RBC Transfusions Over the Course of the Study

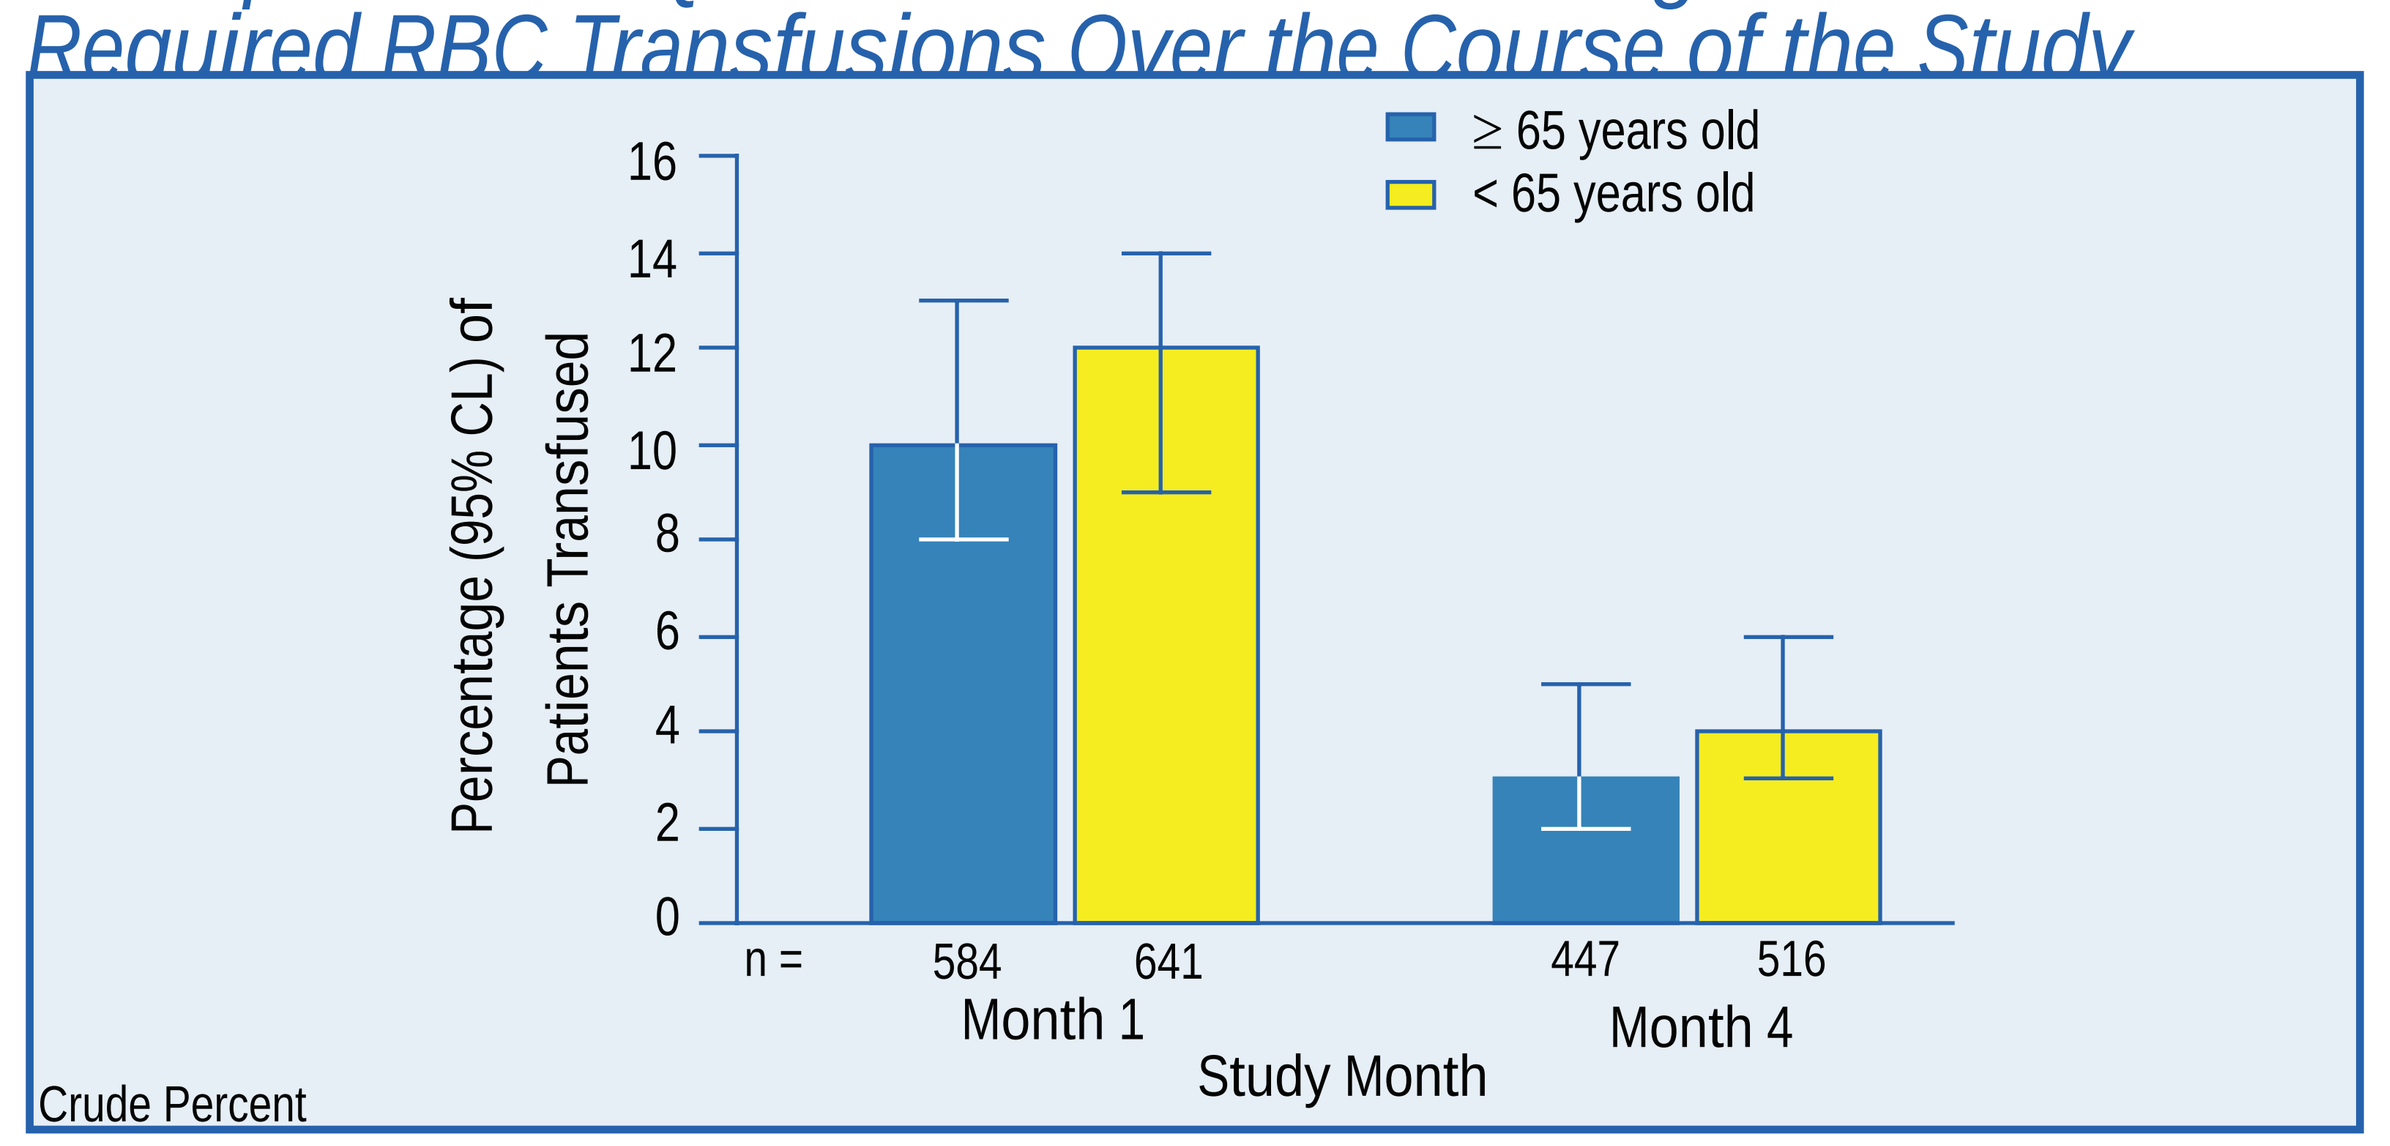
≥ 65 years old
< 65 years old
Percentage (95% CL) of Patients Transfused
16
14
12
10
8
6
4
2
0
n =
584
641
447
516
Month 1
Month 4
Study Month
Crude Percent

Quality of Life Endpoints

	Elderly Patients (≥ 65 years old) (n = 584)	Younger Patients (< 65 years old) (n = 641)
Mean (95% CL) change in FACT-F from baseline to week 7 [n]	0.0 (-1.0, 1.1) [473]	2.6 (1.6, 3.6) [546]
Mean (95% CL) change in FACT-F from baseline to week 16 [n]	2.6 (1.3, 4.0) [392]	6.4 (5.1, 7.7) [444]

Available data. Change in FACT-F ≥ 3 points has been reported to be clinically significant[12]

Darbepoetin alfa Utilization

	Elderly Patients (≥ 65 years old) (n = 584)	Younger Patients (< 65 years old) (n = 641)
Mean (95% CL) weeks of dosing [n]	10.9 (10.6, 11.2)	11.4 (11.1, 11.6)
Mean (95% CL) average Q3W dose administered (mcg/dose)	321.7 (317.2, 326.1)	326.3 (321.9, 330.8)
Mean (95% CL) number of doses received	4.3 (4.1, 4.3)	4.3 (4.2, 4.4)
Number of patients with at least 1 dose withheld – n (%)	81 (14)	107 (17)
Number of patients who had a dose increase – n (%)	201 (34)	245 (38)

SAFETY

Proportion of Patients with Serious Adverse Events

	Elderly Patients (≥ 65 years old) (n = 584)	Younger Patients (< 65 years old) (n = 641)
Subjects with serious adverse events - n (%)	172 (29%)	154 (24%)
Subjects with treatment-related serious adverse events - n (%)	4 (1%)	6 (1%)

- Treatment-related serious adverse events included, deep vein thrombosis in 2 patients (< 1%) and pulmonary emboli in 1 patient (< 1%) from each age strata
- At the time of this interim analysis, no patients had neutralizing antibodies to darbepoetin alfa

CONCLUSIONS

- Darbepoetin alfa 300 mcg Q3W is well tolerated in elderly (≥ 65 years old) and younger (< 65 years old) cancer patients with CIA
- Elderly and younger patients experienced similar clinical outcomes with respect to achievement and maintenance of Hb levels and reduction in the incidence of RBC transfusions
- However, younger patients experienced greater improvement in symptoms of fatigue (measured using FACT-F) than elderly patients
- The dosing regimen of Q3W may simplify the treatment of CIA in the oncology practice by synchronizing darbepoetin alfa therapy with chemotherapy administered Q3W, thereby minimizing disruption to the lives of patients and their caregivers

AMGEN®